Deucravacitinib exposure-response analyses in patients with moderate to severe psoriasis

34507

Jun Shen,[1] Mark Lovern,[2] Matt Maloney,[1] Amit Roy,[1] Elizabeth Colston,[1] Anjaneya Chimalakonda[1]

[1]Bristol Myers Squibb, Princeton, NJ, USA; [2]Certara, Princeton, NJ, USA

Introduction

- Tyrosine kinase 2 (TYK2) is an intracellular enzyme required for signal transduction and cellular functions downstream of Type I interferons and interleukin-23, which are involved in the initiation and maintenance of psoriatic disease and other immune-mediated disorders[1]
- Deucravacitinib is an oral, allosteric inhibitor of TYK2 that achieves high selectivity by uniquely binding to the regulatory domain of the enzyme rather than to the conserved active domain where other kinase inhibitors bind, including Janus kinase (JAK) 1/2/3 inhibitors[1,2]
 - The high functional selectivity of deucravacitinib for TYK2 may provide a differentiated risk-benefit profile compared with JAK 1/2/3 inhibitors due to potent TYK2 inhibition and minimal activity against JAK 1/2/3[1,2]

Objective

- To evaluate the relationships of deucravacitinib exposure with efficacy and safety in patients with moderate to severe plaque psoriasis

Methods

- Exposure-response (E-R) analyses included patients from the phase 2, randomized, double-blind psoriasis trial (NCT02931838)[3] and the phase 3, randomized, double-blind POETYK PSO-1 (NCT03624127) and PSO-2 (NCT03611751) trials[4]
 - POETYK PSO-1 and PSO-2 were 52-week trials, with the coprimary endpoints (≥75% reduction from baseline in Psoriasis Area and Severity Index [PASI 75] and static Physician's Global Assessment [sPGA] score of 0/1) assessed at Week 16[4]
 - In the phase 2 trial, the primary endpoint (PASI 75) was assessed at Week 12[3]
- Dose groups included placebo and deucravacitinib 3 mg every other day, 3 mg once daily (QD), 3 mg twice daily (BID), 6 mg QD, 6 mg BID, and 12 mg QD
- Efficacy endpoints included in this analysis were ≥75%/≥90%/100% reduction from baseline in PASI (PASI 75/90/100) and sPGA 0/1 responses
- Safety endpoints included major adverse cardiovascular events (MACE), extended MACE, malignancies, overall infections and infestations, herpes zoster infections, serious infections, and creatine phosphokinase (CPK) elevations from baseline of grade ≥3
- E-R analyses of efficacy endpoints were described by multivariable longitudinal proportional-odds ordered categorical models, and E-R analysis of infections/infestations (only safety endpoint with sufficient events to merit such analysis) was described by a logistic regression model
 - Covariates included patient age, race, sex, body weight, smoking status, geographic region, baseline PASI score, baseline body surface area (BSA) involvement, and prior biologic therapy use
- Additionally, exposure-quartile plots of the incidence of infections and infestations and CPK elevations of grade ≥3 at Week 12/16 and Week 52 were developed

Results

Data analyses across the phase 2 and phase 3 trials

- A total of 1524 and 1522 patients were included in the E-R analyses of PASI and sPGA, respectively, across the 3 trials (**Table 1**)
 - 1524 patients were included in the E-R safety analysis

Table 1. Patients included in the E-R analysis of efficacy and safety

Trial	Placebo or deucravacitinib-treated patients, n	Patients included in E-R analysis, n (%)
PASI[a]		
Phase 2	267	267 (100)
POETYK PSO-1	497	496 (99.8)
POETYK PSO-2	764	761 (99.6)
Total	**1528**	**1524 (99.7)**
sPGA[a]		
Phase 2	267	265 (99.3)
POETYK PSO-1	497	496 (99.8)
POETYK PSO-2	764	761 (99.6)
Total	**1528**	**1522 (99.6)**
Safety[b]		
Phase 2	267	267 (100)
POETYK PSO-1	497	496 (99.8)
POETYK PSO-2	764	761 (99.6)
Total	**1528**	**1524 (99.7)**

[a]The E-R efficacy analysis population included patients who received ≥1 dose of deucravacitinib or placebo, had deucravacitinib exposure available, and had ≥1 baseline and ≥1 postbaseline PASI or sPGA response value available. Drug exposures were imputed to be zero for patients who received placebo.
[b]The E-R safety analysis population included patients who received ≥1 dose of deucravacitinib or placebo and had deucravacitinib exposure available. Drug exposures were imputed to be zero for patients who received placebo.
E-R, exposure-response; PASI, Psoriasis Area and Severity Index; sPGA, static Physician's Global Assessment.

E-R analyses of efficacy across the phase 2 and 3 trials

- E-R analyses demonstrated an exposure-dependent increase in PASI 75 and sPGA 0/1 responses (**Figure 1**)
- A 6 mg QD dose provided near-maximal efficacy, considering that the model-predicted probability of PASI 75 and sPGA 0/1 responses at Week 16 and Week 24 for the 6 mg QD dose was close to maximal response
 - A 2- to 3-fold increase in exposure vs the deucravacitinib 6 mg QD dose was predicted to slightly (~4%–7%) increase PASI 75 or sPGA 0/1 responses, indicating that the 6 mg QD dose achieved close to maximal response (data not shown)

Figure 1. Predicted median (90% PI) probability of PASI 75 and sPGA 0/1 responses at Weeks 16 and 24 vs time-averaged deucravacitinib steady-state concentration (placebo and deucravacitinib-treated patients in phase 2 and phase 3 trials)

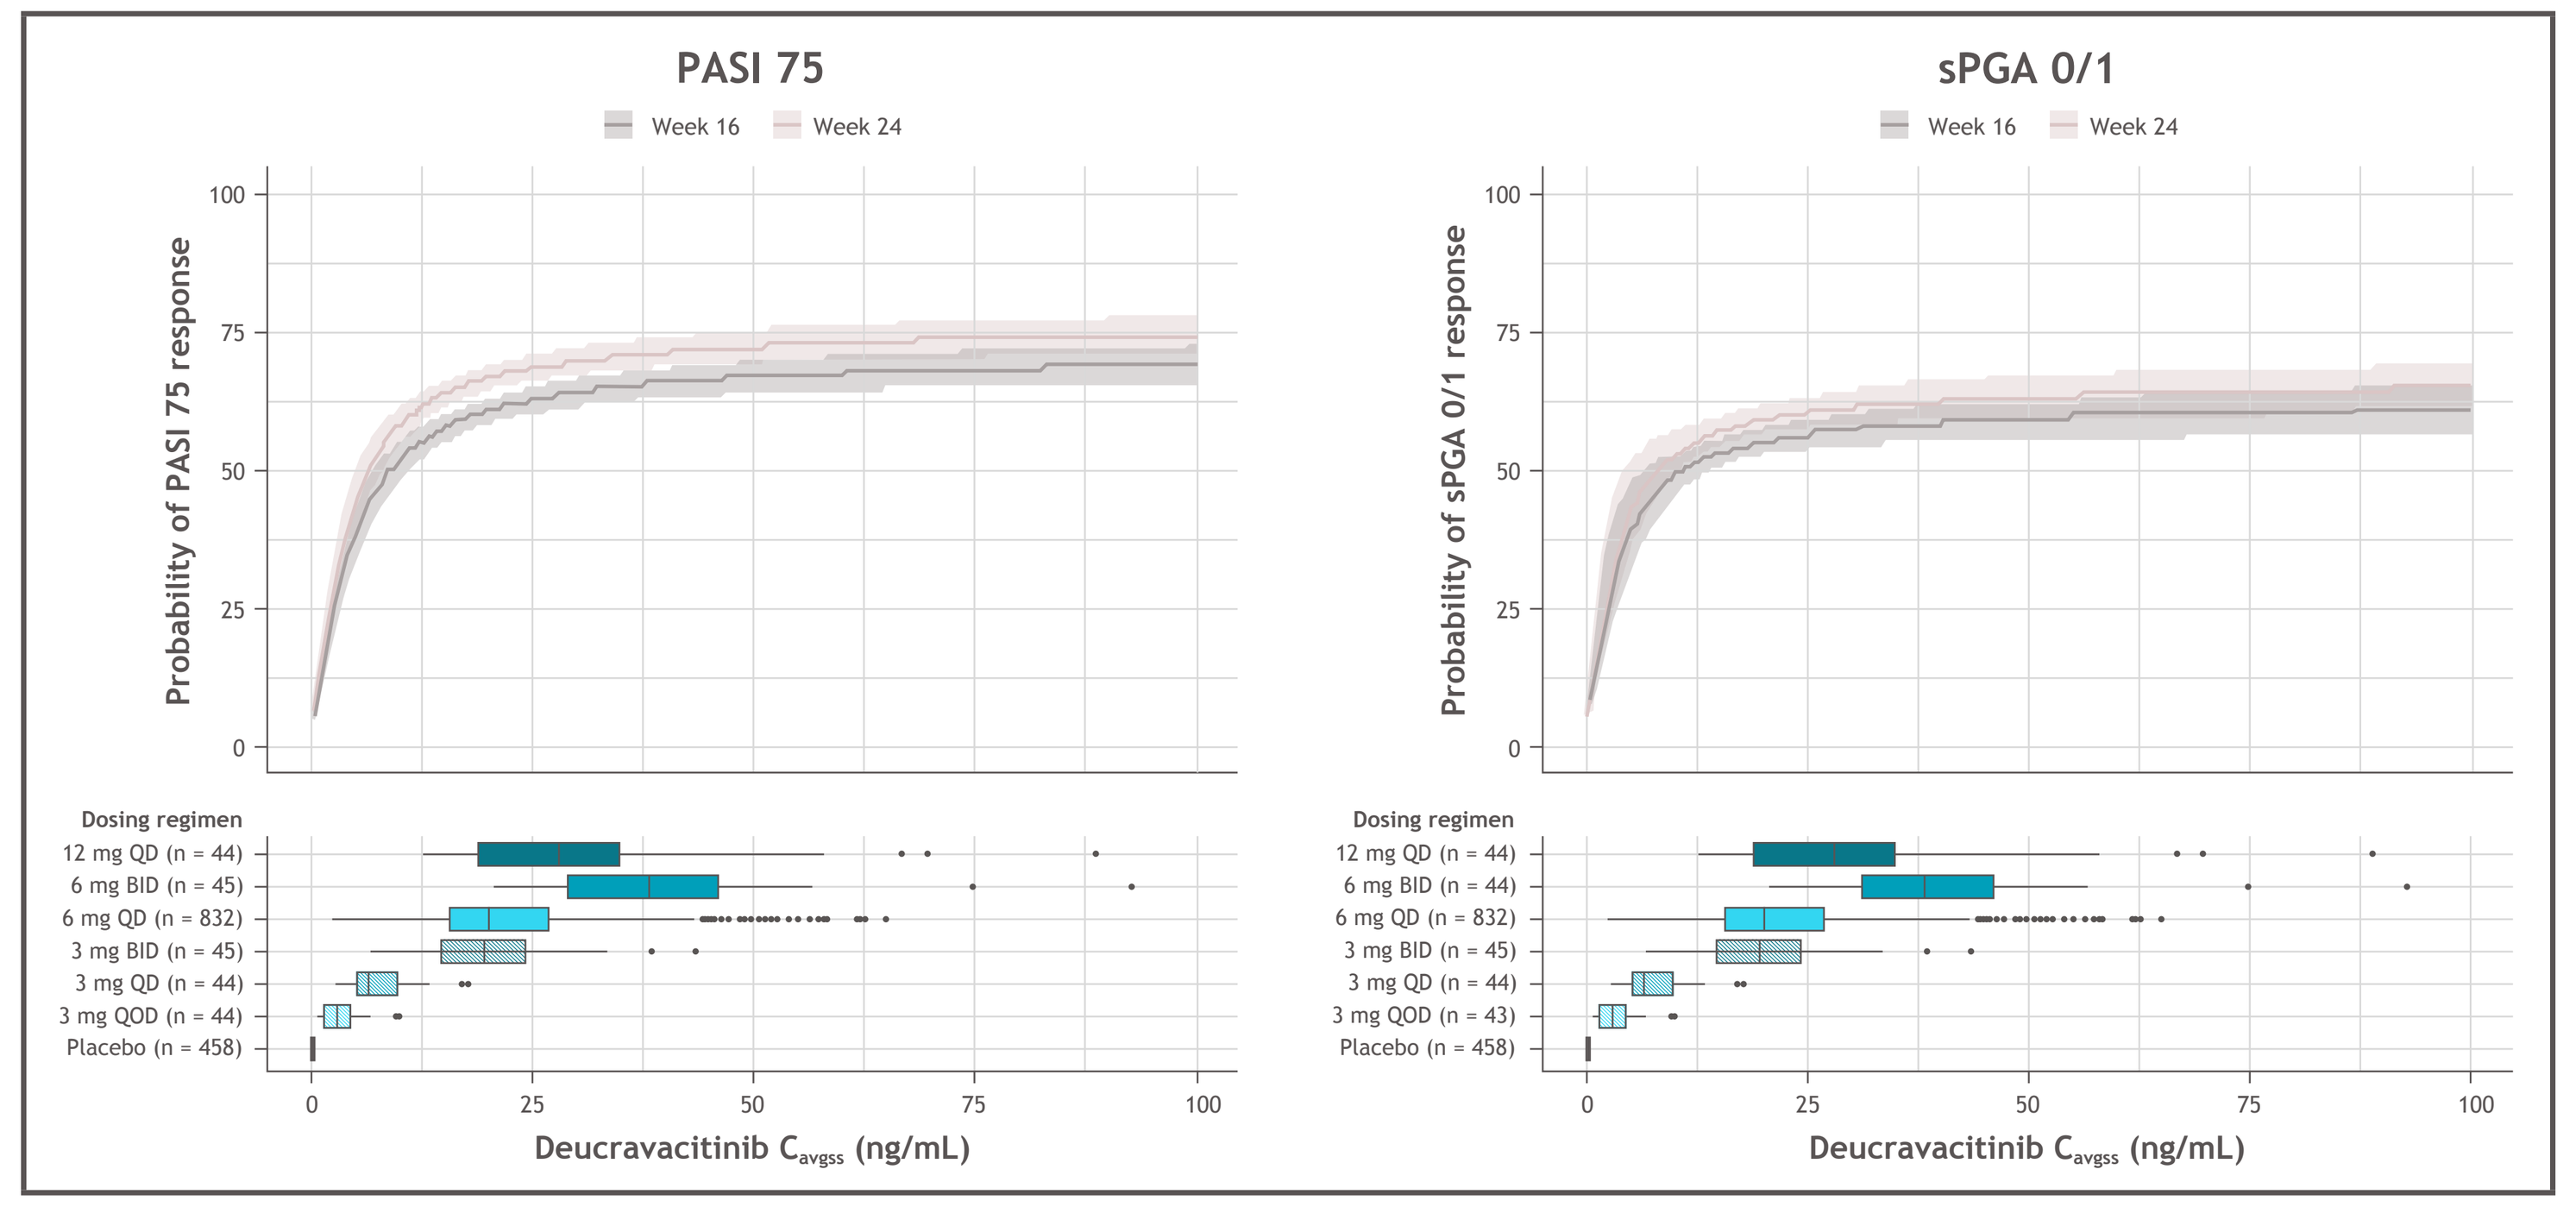
PASI 75
sPGA 0/1
Week 16
Week 24
Probability of PASI 75 response
Probability of sPGA 0/1 response
Dosing regimen
12 mg QD (n = 44)
6 mg BID (n = 45)
6 mg QD (n = 832)
3 mg BID (n = 45)
3 mg QD (n = 44)
3 mg QOD (n = 44)
Placebo (n = 458)
Deucravacitinib C_{avgss} (ng/mL)

The solid curve on the upper panel gives the model-predicted median PASI 75 or sPGA 0/1 probability for Week 16 or Week 24 with a ribbon showing the corresponding 90% PI. The boxplots at the bottom represent the model-predicted exposure range achieved by each dosing regimen.
BID, twice daily; C_{avgss}, average plasma drug concentration at steady state; PASI 75, ≥75% reduction from baseline in Psoriasis Area and Severity Index; PI, prediction interval; QD, once daily; QOD, every other day; sPGA, static Physician's Global Assessment.

- Model predictions indicated that a 6 mg QD dose provided close to maximal PASI 75 and sPGA 0/1 responses at Weeks 16 and 24 across relevant subgroups based on patient age, sex, body weight, and previous biologic use
- In patients receiving deucravacitinib 6 mg QD, the PASI model captured onset, peak, and continuation of PASI 75/90/100 response rates (**Figure 2**)

Figure 2. Observed and predicted (median, 90% PI) PASI 75/90/100 responses over time in patients with psoriasis treated with deucravacitinib 6 mg QD

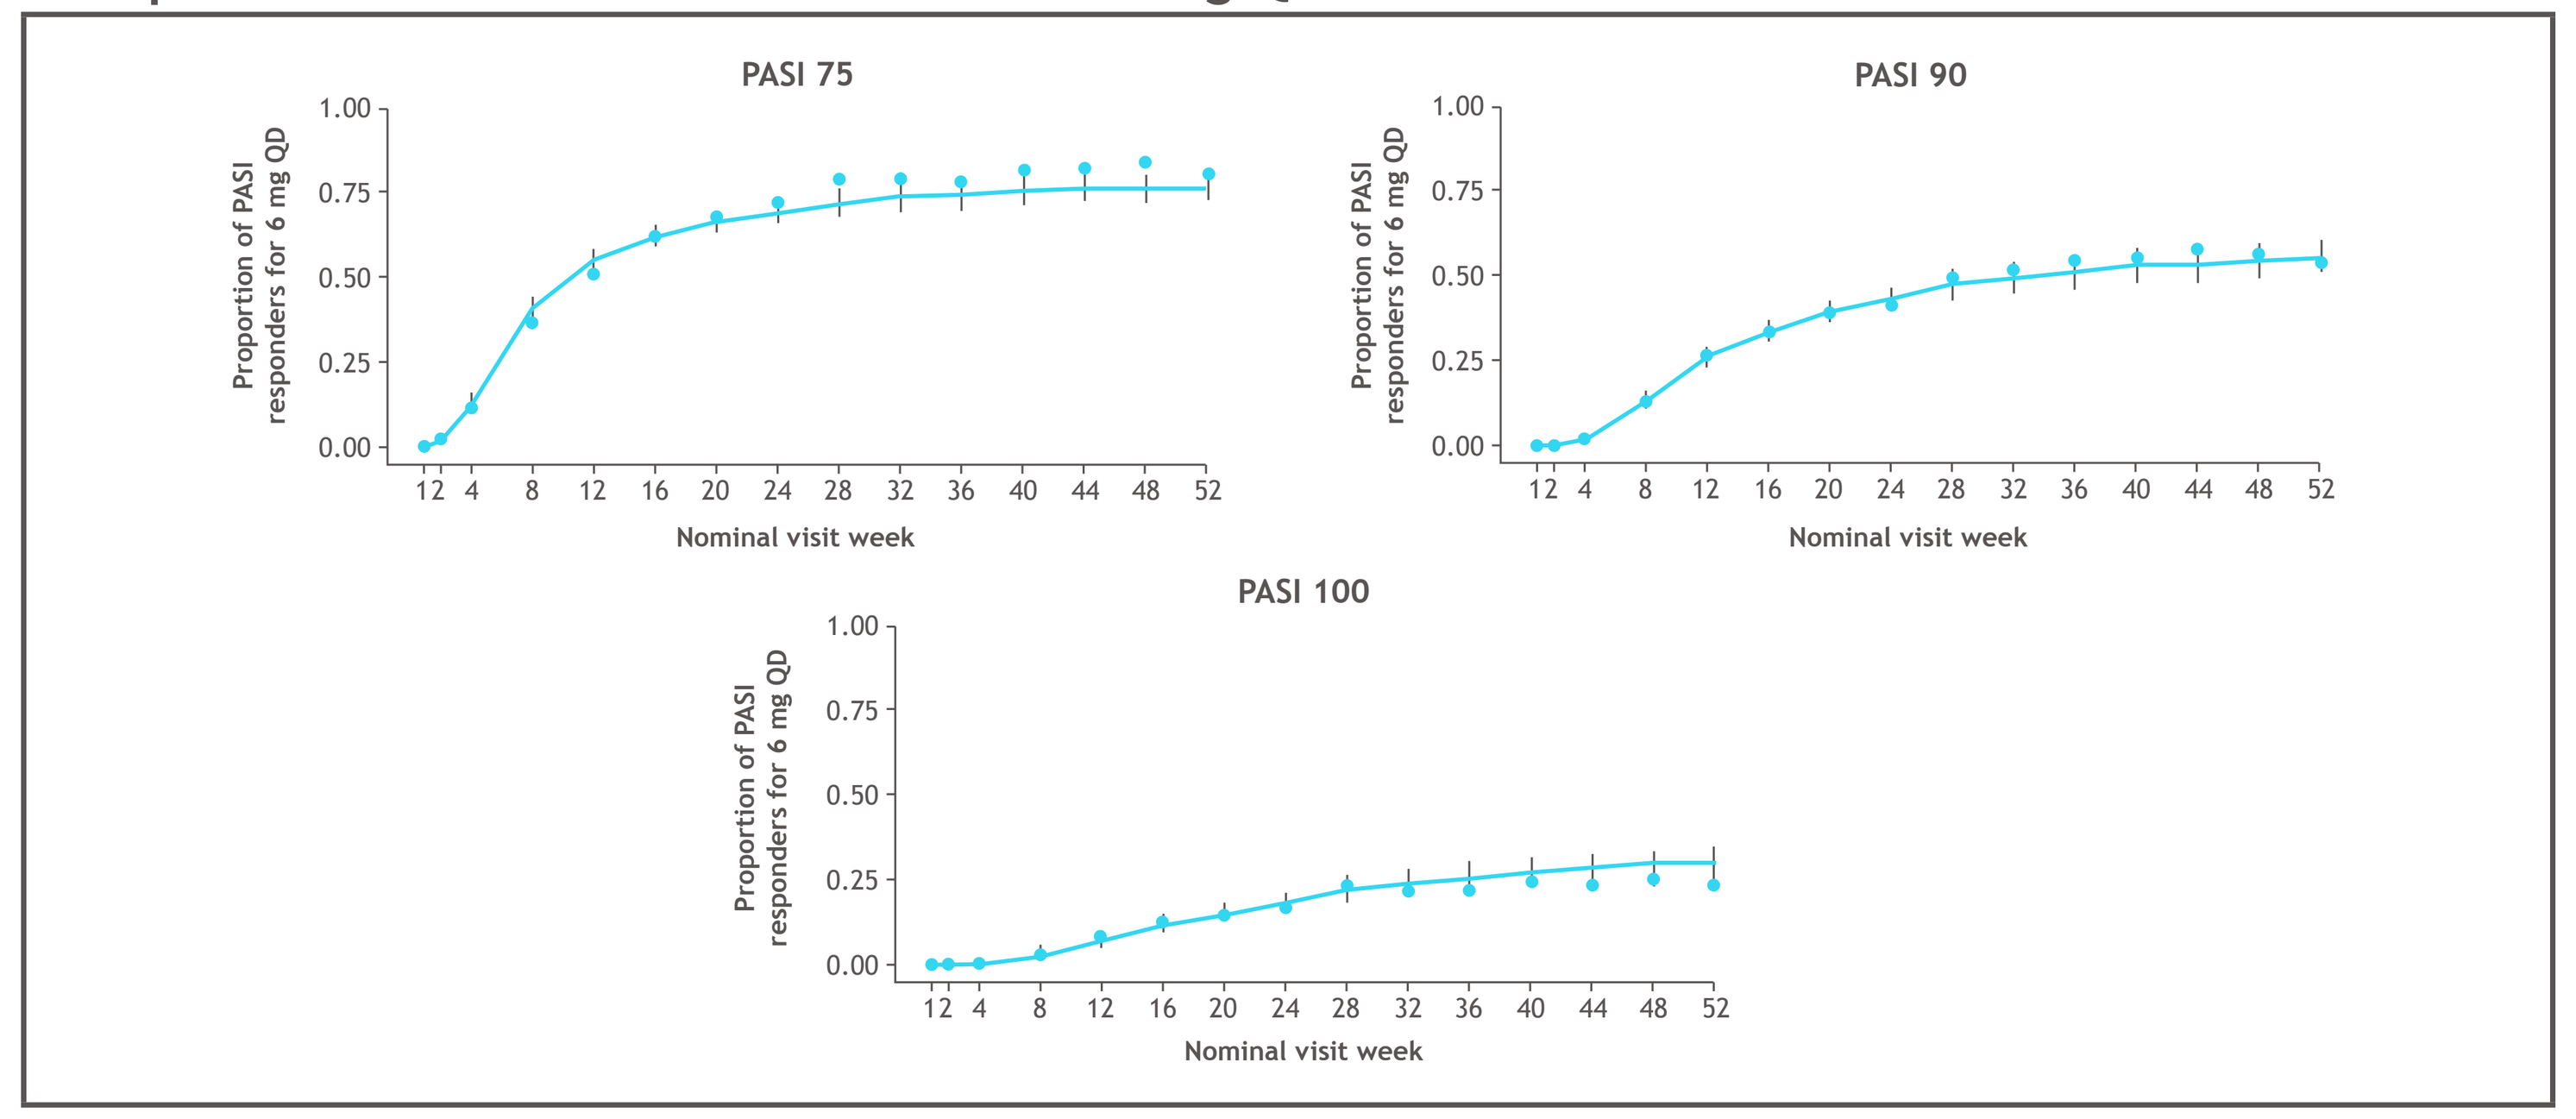
PASI 75
PASI 90
PASI 100
Proportion of PASI responders for 6 mg QD
Nominal visit week

The blue dots represent the observed response rate at a specific nominal visit week, the blue lines represent the median model-predicted response rate, and the black vertical lines represent the 90% PI for a specific visit week.
PASI 75/90/100, ≥75%/≥90%/100% reduction from baseline in Psoriasis Area and Severity Index; PI, prediction interval; QD, once daily.

- In patients receiving deucravacitinib 6 mg QD, the sPGA E-R model captured onset, peak, and continuation of sPGA 0 and sPGA 0/1 responses (**Figure 3**)

Figure 3. Observed and predicted (median, 90% PI) sPGA 0 and sPGA 0/1 responses over time in patients with psoriasis treated with deucravacitinib 6 mg QD

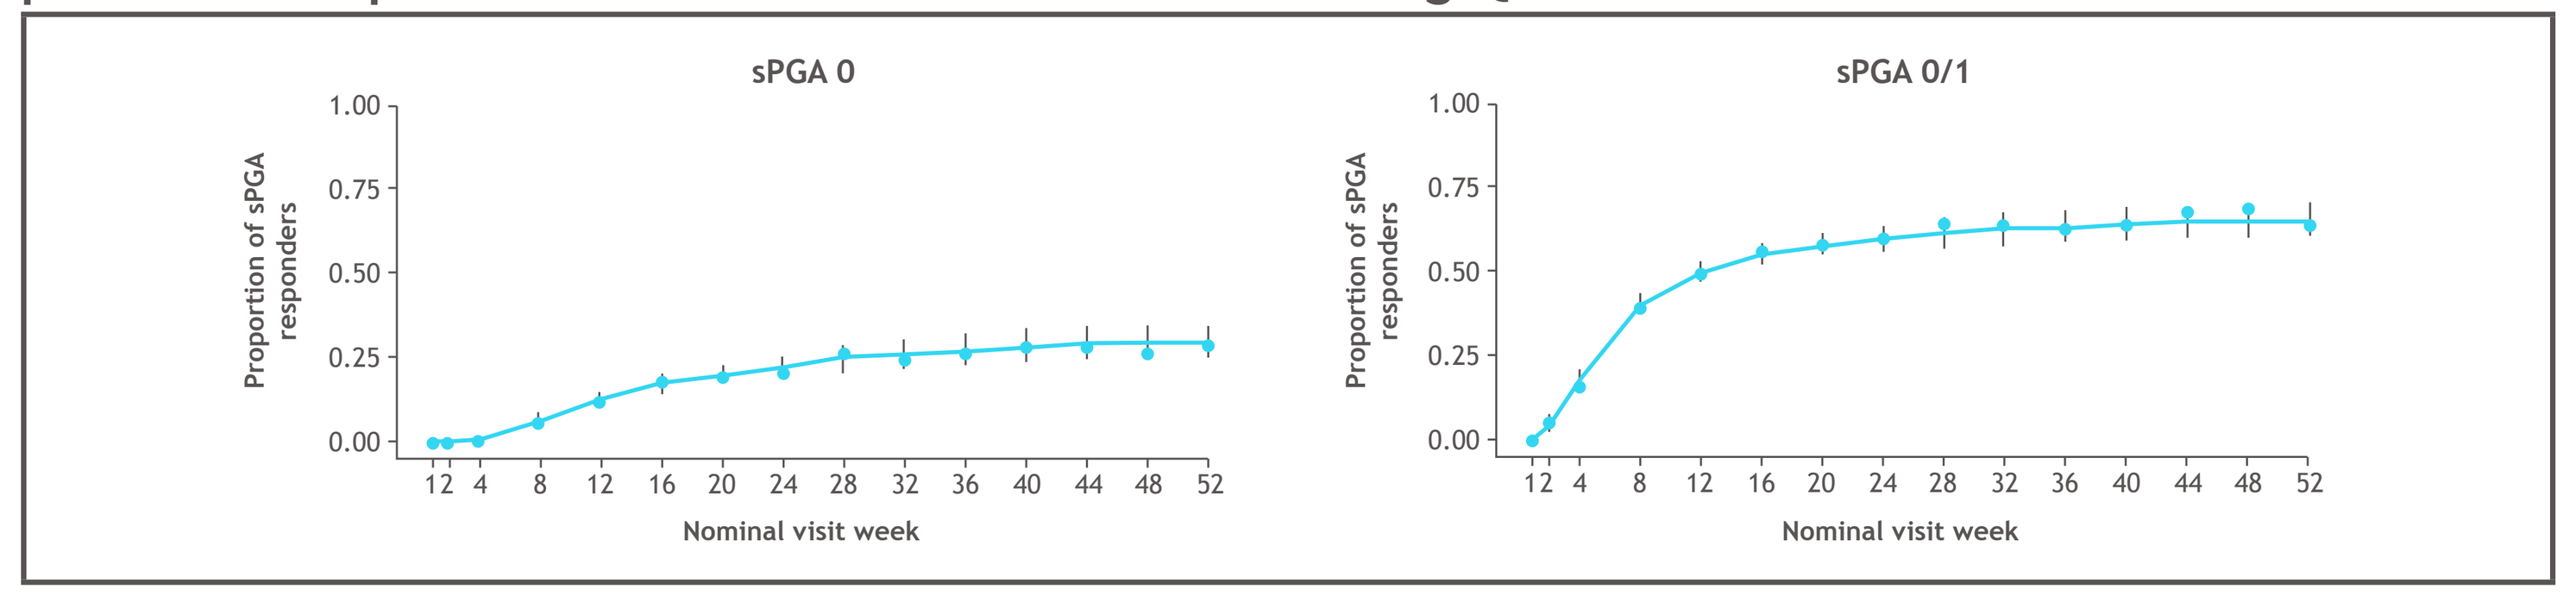
sPGA 0
sPGA 0/1
Proportion of sPGA responders
Nominal visit week

The blue dots show the response rate at a specific nominal visit week, the blue lines represent the median model-predicted response rate, and the black vertical lines represent the 90% PI.
PI, prediction interval; QD, once daily; sPGA, static Physician's Global Assessment.

E-R analyses of safety across the phase 2 and 3 trials

- No E-R trends were observed for MACE, extended MACE, serious infections, herpes zoster infections, or malignancies at Week 12/16 (**Figure 4**) or at Week 52 (**Figure 5**)
 - Incidences of these adverse events were relatively low (<10%) in patients originally randomized to deucravacitinib and treated with deucravacitinib up to Week 12/16 or Week 52
- No E-R trends were observed between deucravacitinib exposure and the occurrence of grade ≥3 CPK elevations at Week 12/16 and Week 52 (**Figure 6**)

Figure 4. Distribution of deucravacitinib concentration by safety events in the phase 2 and phase 3 trials (Week 12 [phase 2 trial] or Week 16 [phase 3 trials])

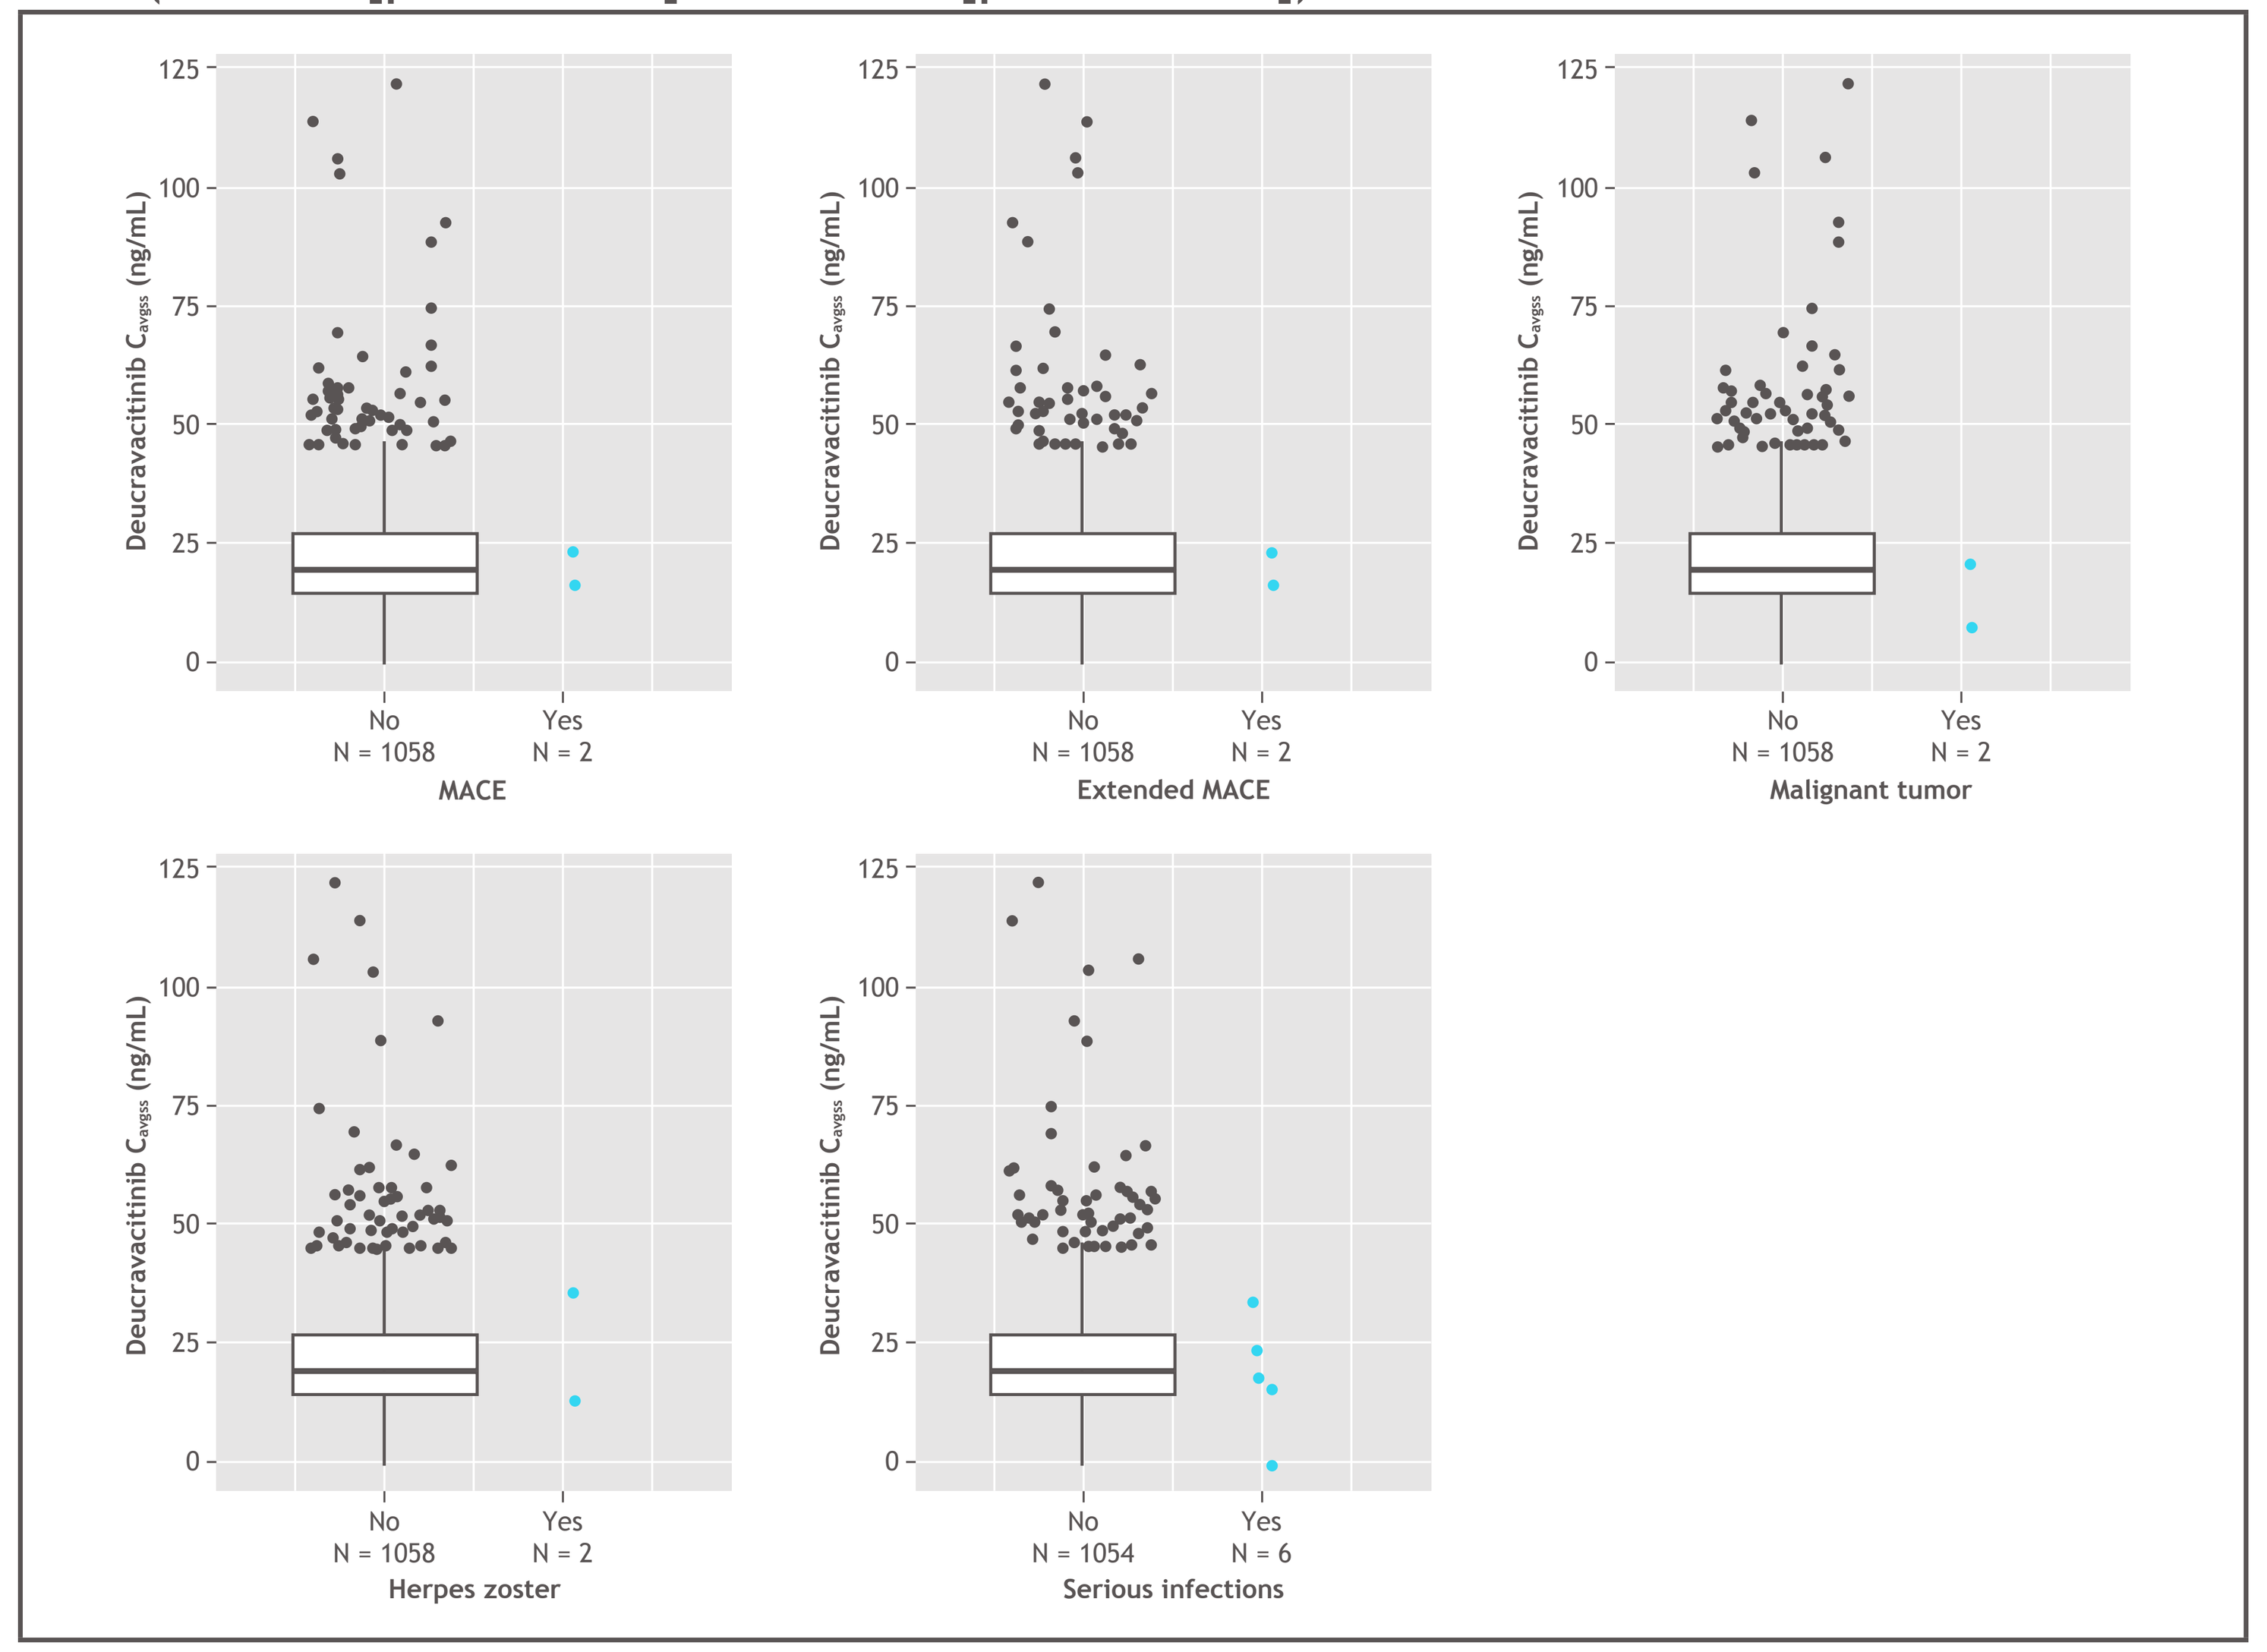
Deucravacitinib C_{avgss} (ng/mL)
No N = 1058
Yes N = 2
MACE
No N = 1058
Yes N = 2
Extended MACE
No N = 1058
Yes N = 2
Malignant tumor
No N = 1058
Yes N = 2
Herpes zoster
No N = 1054
Yes N = 6
Serious infections

Adverse events are included in this analysis for patients who were originally randomized to deucravacitinib treatment in the phase 2/3 trials. Primary endpoints were assessed at Week 12 in the phase 2 trial and at Week 16 in the phase 3 trials.
C_{avgss}, average plasma drug concentration at steady state; MACE, major adverse cardiovascular events.

Figure 5. Distribution of deucravacitinib concentration by safety events in the phase 3 trials (Week 52)

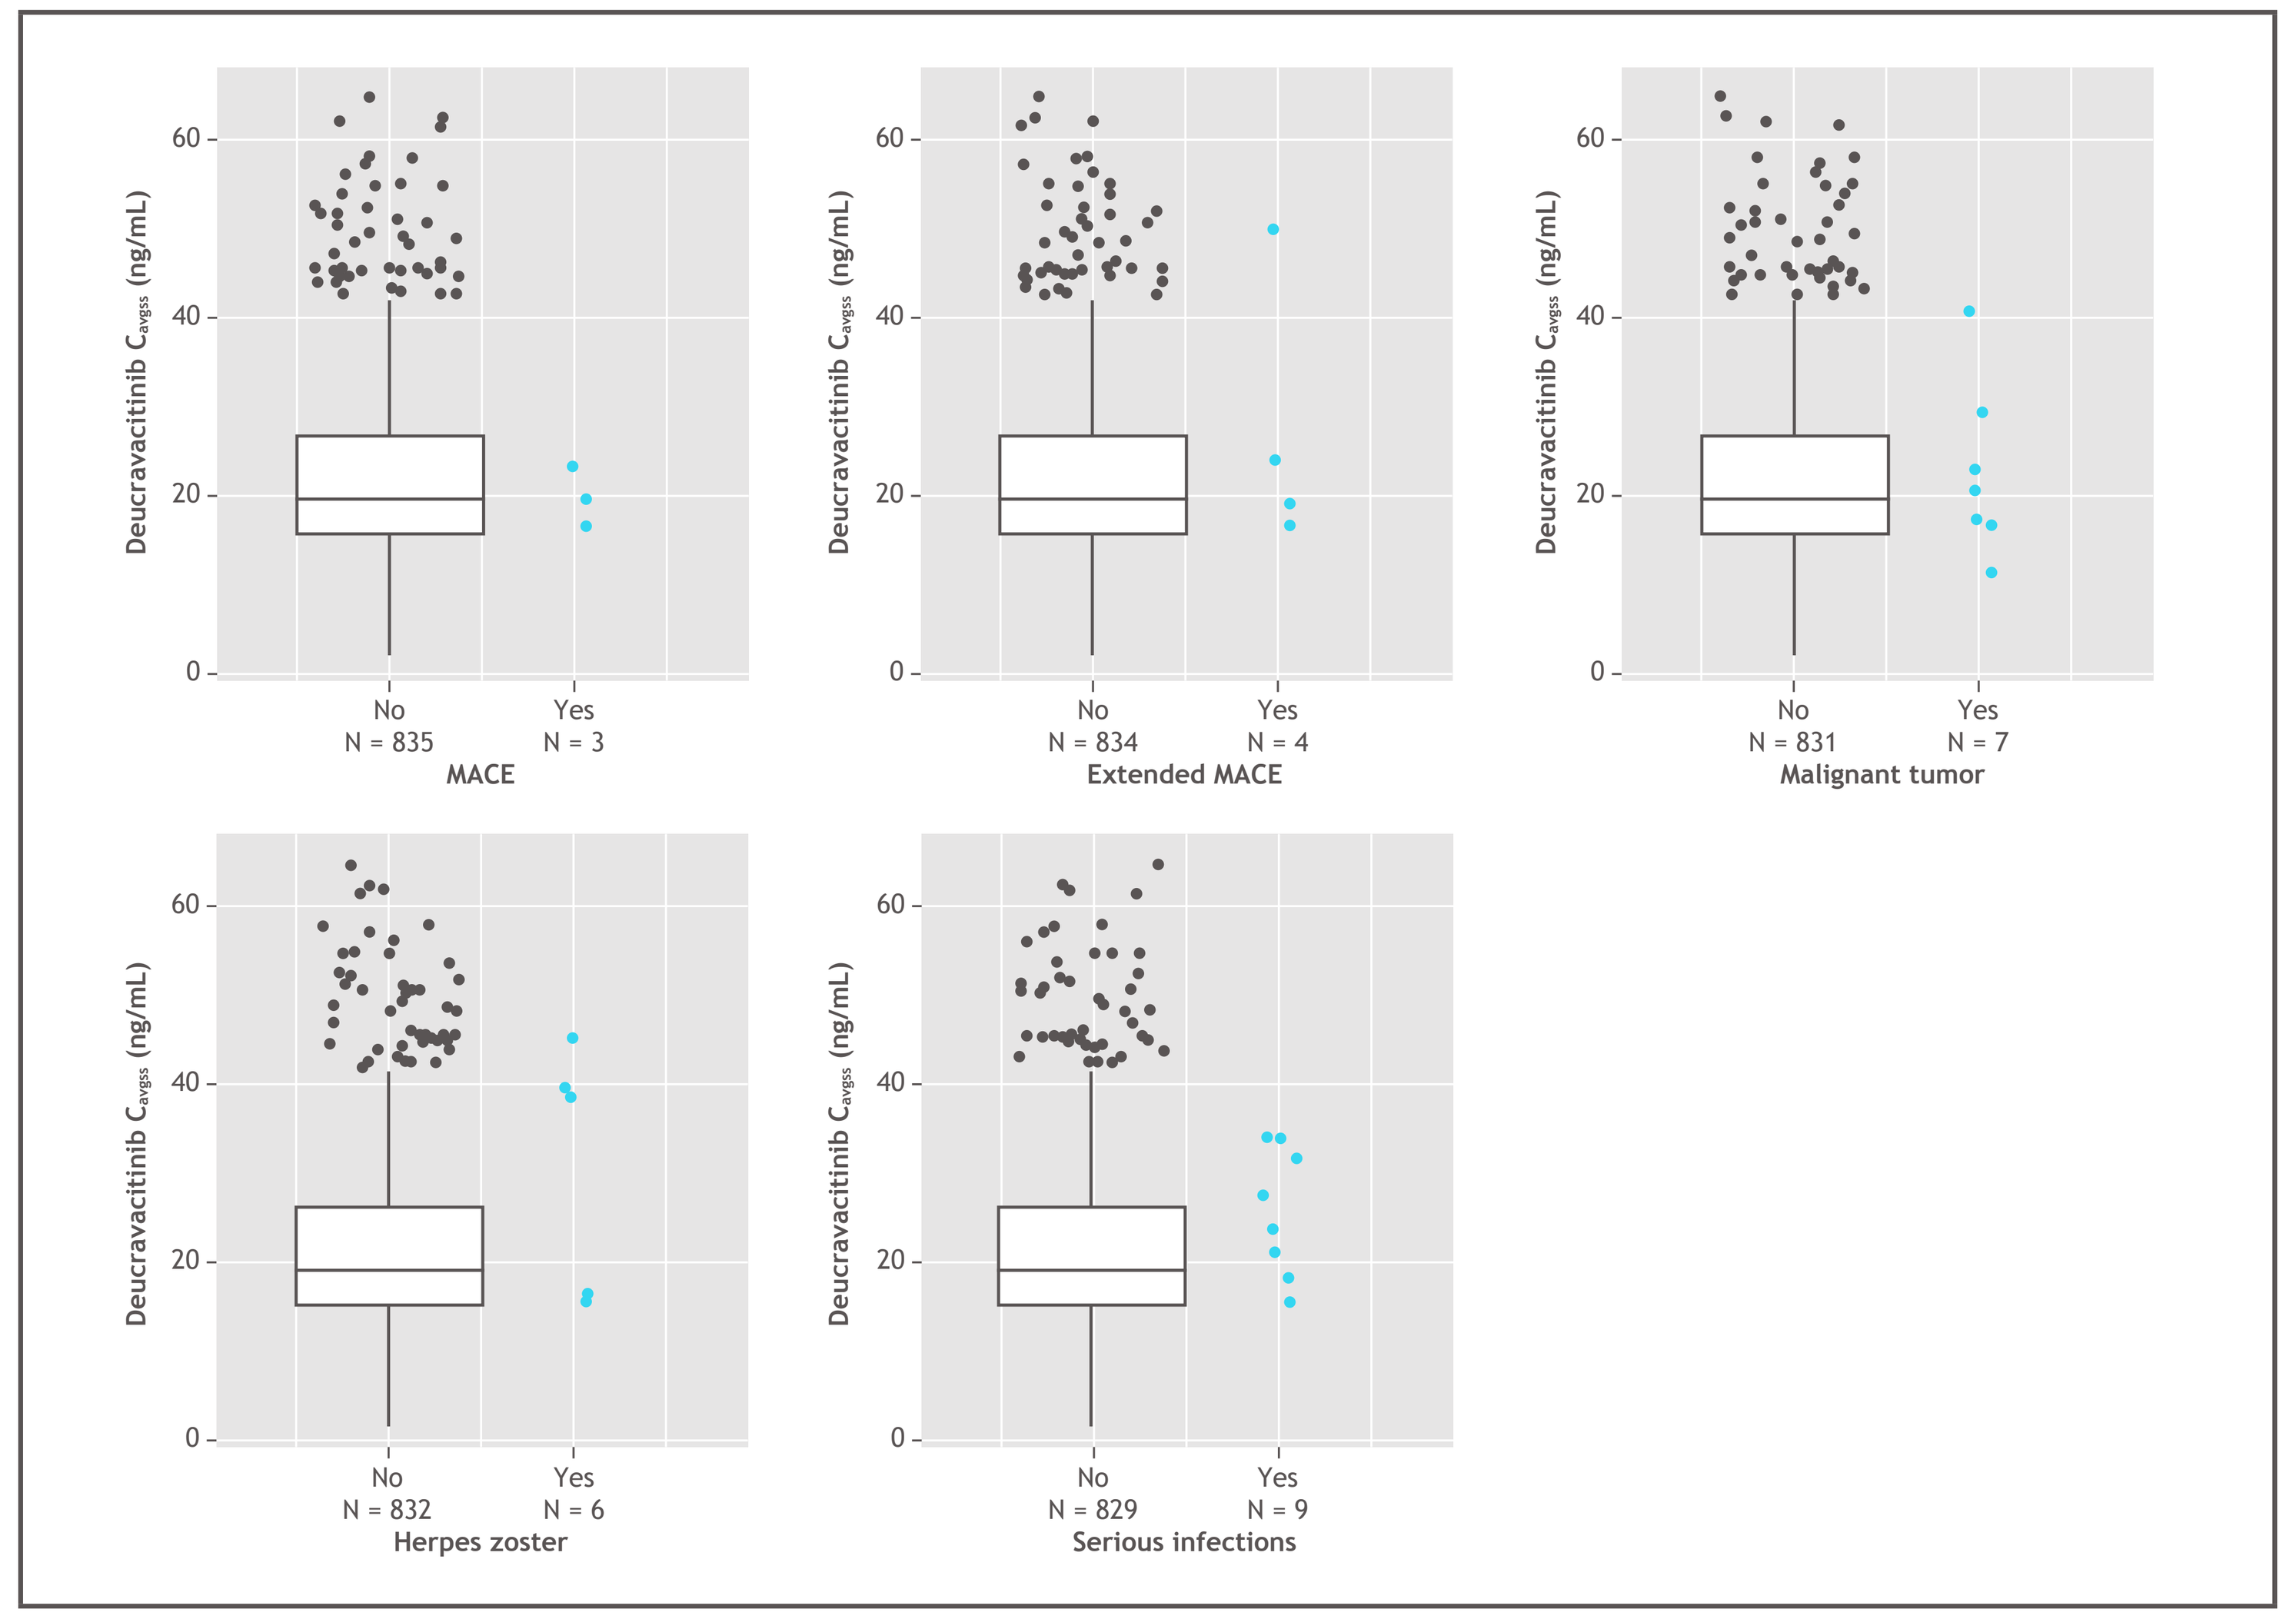
Deucravacitinib C_{avgss} (ng/mL)
No N = 835
Yes N = 3
MACE
No N = 834
Yes N = 4
Extended MACE
No N = 831
Yes N = 7
Malignant tumor
No N = 832
Yes N = 6
Herpes zoster
No N = 829
Yes N = 9
Serious infections

Adverse events are included in this analysis for patients who were originally randomized to deucravacitinib treatment in the phase 3 trials. Events occurring in patients who switched treatment during the trials were not included in this analysis.
C_{avgss}, average plasma drug concentration at steady state; MACE, major adverse cardiovascular events.

Figure 6. E-R quartile plots for CPK at Week 12/16 and Week 52

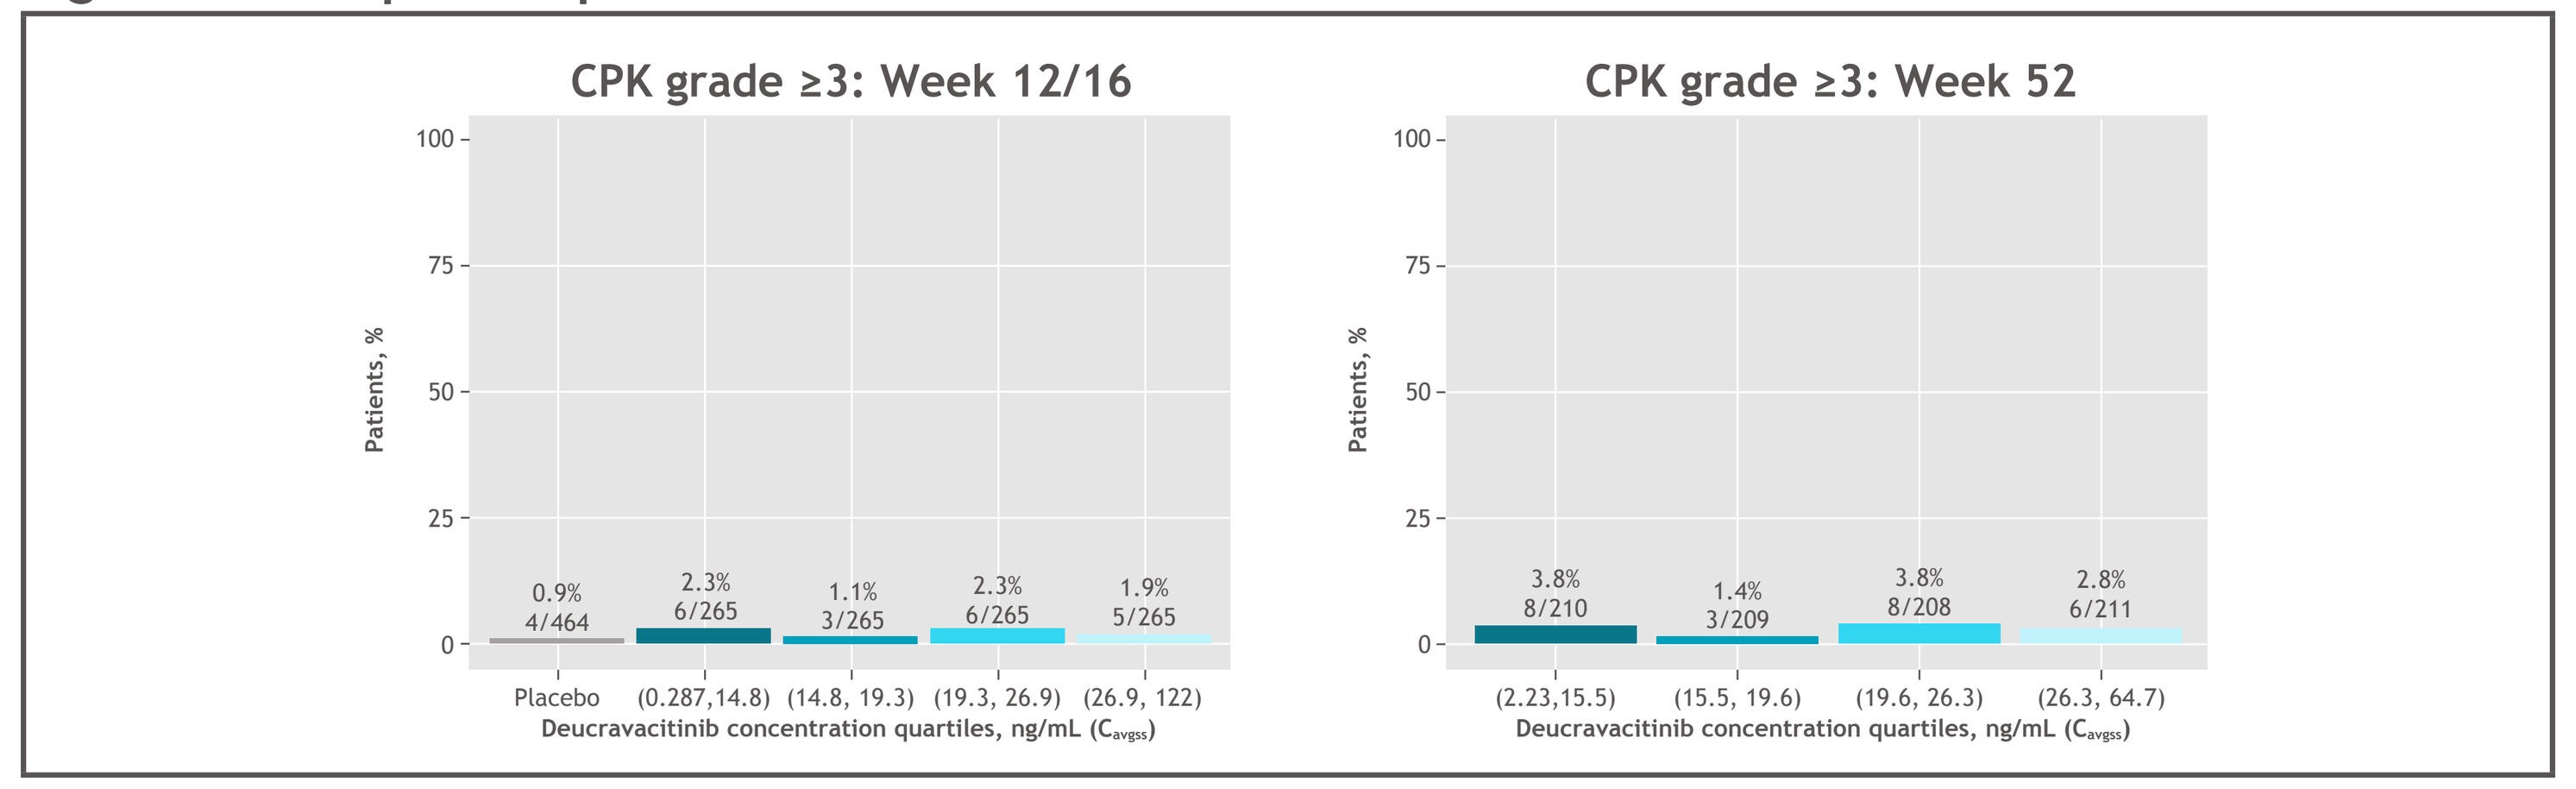
CPK grade ≥3: Week 12/16
CPK grade ≥3: Week 52
Patients, %
0.9% 4/464
2.3% 6/265
1.1% 3/265
2.3% 6/265
1.9% 5/265
Placebo
(0.287,14.8)
(14.8, 19.3)
(19.3, 26.9)
(26.9, 122)
3.8% 8/210
1.4% 3/209
3.8% 8/208
2.8% 6/211
(2.23,15.5)
(15.5, 19.6)
(19.6, 26.3)
(26.3, 64.7)
Deucravacitinib concentration quartiles, ng/mL (C_{avgss})

Week 12/16 plots include adverse events for patients originally randomized to deucravacitinib treatment or placebo in the phase 2/3 trials for the period during which the patient did not switch treatment. Events occurring after patients switched treatment are not included in this analysis.
Week 52 plots include adverse events only for patients originally randomized to deucravacitinib treatment in the phase 3 trials for the period during which the patient did not switch treatment. Events for placebo patients or events occurring after patients switched treatment are not included in this analysis.
Primary endpoints were assessed at Week 12 in the phase 2 trial and at Week 16 in the phase 3 trials.
C_{avgss}, average plasma drug concentration at steady state; CPK, creatine phosphokinase; E-R, exposure-response.

- A relatively flat E-R relationship for infections and infestations with deucravacitinib was apparent at Week 12/16 and was described by a logistic regression model (**Figure 7**); however, an E-R trend for infections and infestations for Weeks 0–52 was not substantial
- A 2-fold increase in deucravacitinib exposure relative to the median exposure of the 6 mg QD dose resulted in a small increase in the risk of infections and infestations (from 29.7% to 32.2%) based on model-predicted data at Week 16
- Given the flat E-R relationship at Week 12/16, an increase in exposure (due to various intrinsic and extrinsic factors) does not result in a clinically meaningful change in probability of infections and infestations

Figure 7. Predicted median (90% PI) probability of infections and infestations vs minimum deucravacitinib plasma concentration (Week 16)

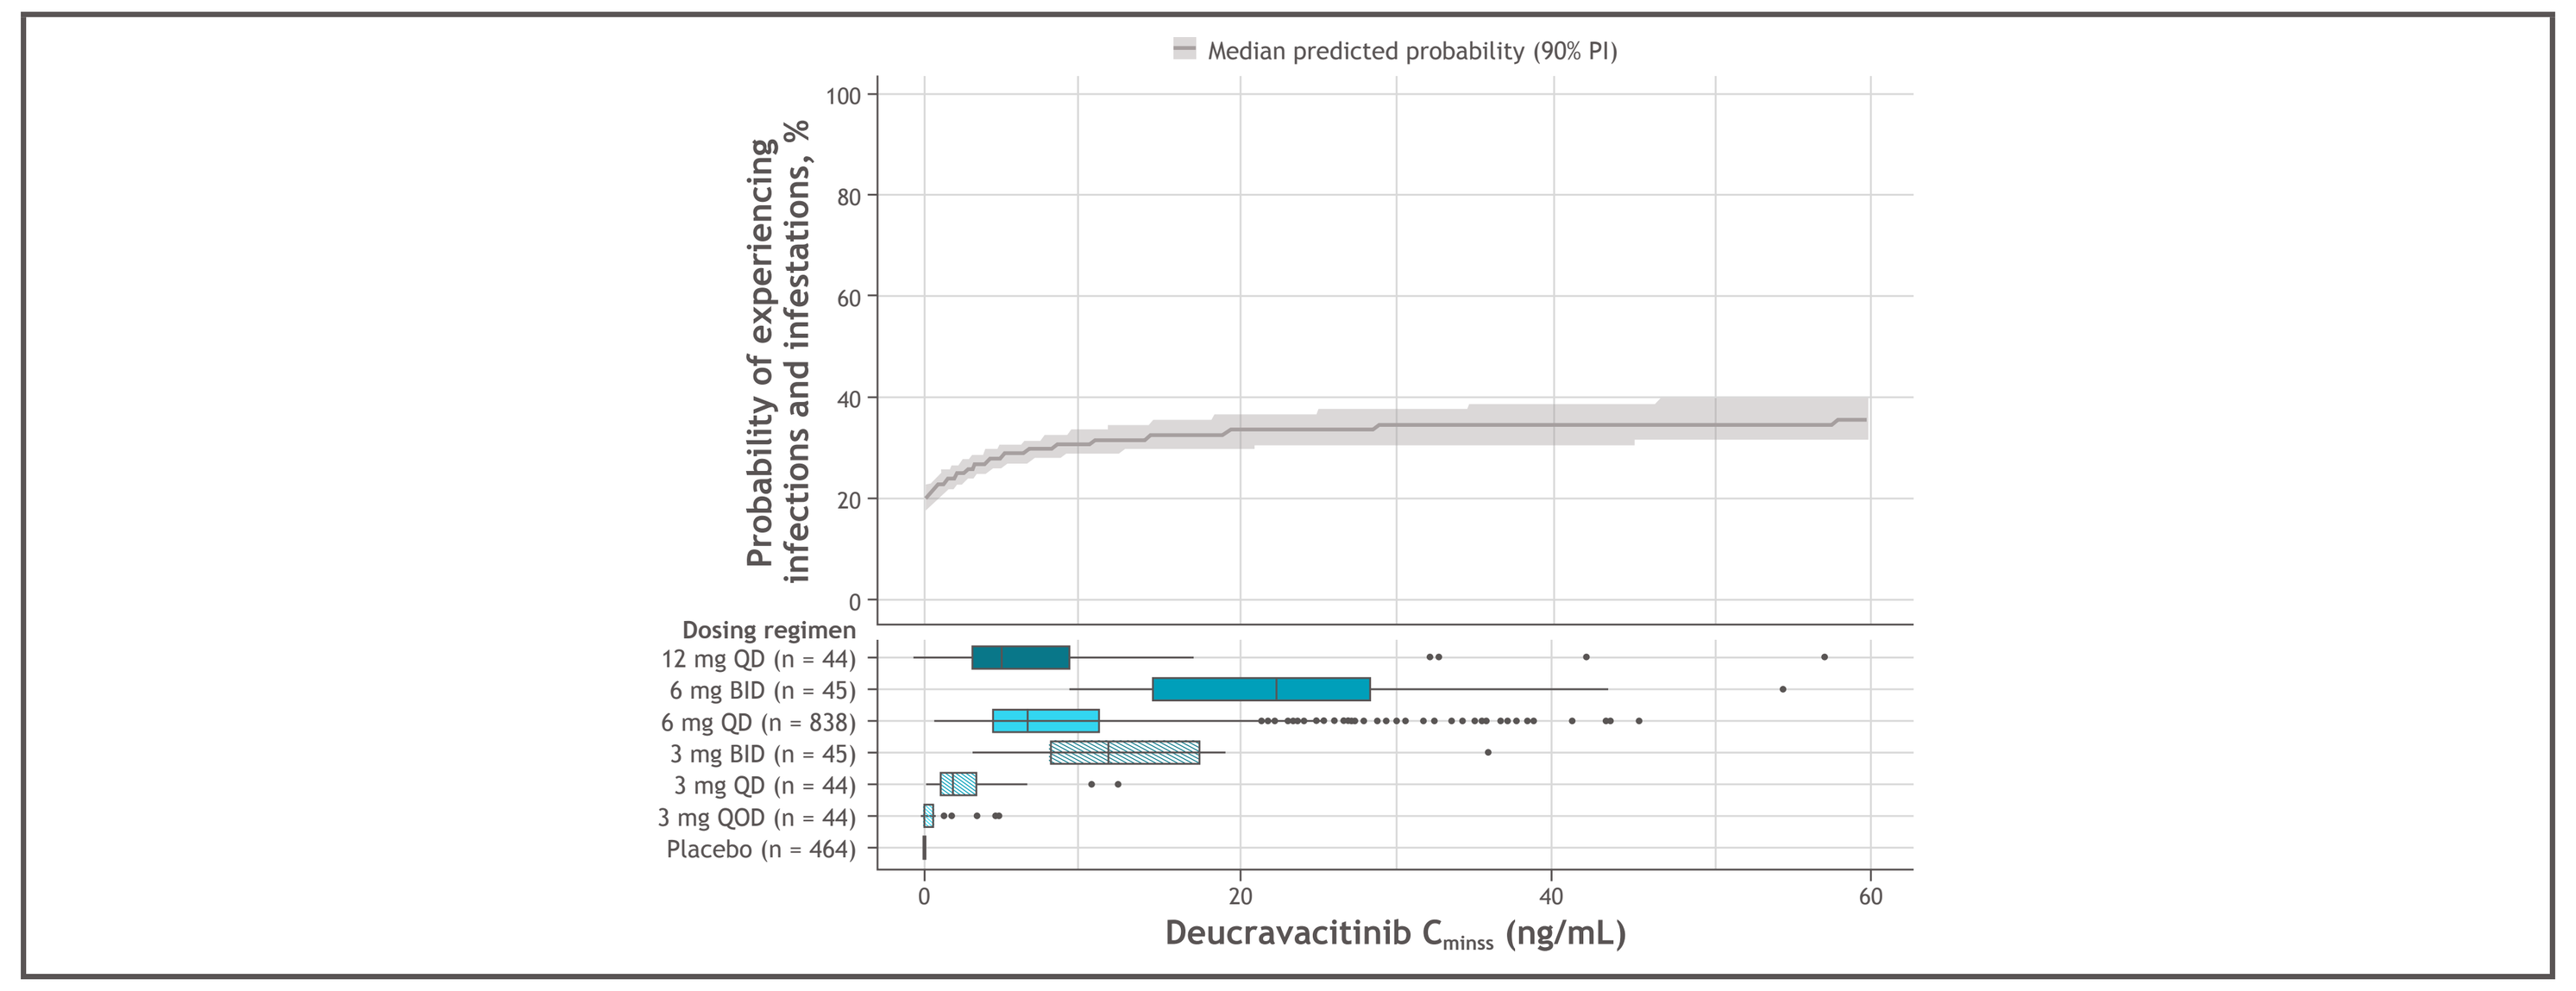
Median predicted probability (90% PI)
Probability of experiencing infections and infestations, %
Dosing regimen
12 mg QD (n = 44)
6 mg BID (n = 45)
6 mg QD (n = 838)
3 mg BID (n = 45)
3 mg QD (n = 44)
3 mg QOD (n = 44)
Placebo (n = 464)
Deucravacitinib C_{minss} (ng/mL)

The solid curve on the upper panel gives the median infections probability, with the ribbon showing the corresponding 90% PI.
BID, twice daily; C_{minss}, minimum plasma drug concentration at steady state; PI, prediction interval; QD, once daily; QOD, every other day.

- The probability of infections and infestations was similar in baseline subgroups based on age, sex, BSA involvement, and prior biologic therapy exposure

Conclusions

- Supporting the clinical superiority of deucravacitinib over placebo, this E-R analysis demonstrated an exposure-dependent increase in PASI 75 and sPGA 0/1 response rates, with the 6 mg QD dose achieving close to maximal response
 - Response rates at the 6 mg QD dose were maintained above placebo in the overall population and across covariate subgroups
- No substantive E-R trends were observed at Weeks 12 and 16 or Week 52 between deucravacitinib exposure and the occurrence of serious infections, herpes zoster infections, MACE, malignancies, or grade ≥3 CPK elevations
 - At Week 16, the overall probability of infections and infestations was modestly greater than placebo at the deucravacitinib 6 mg QD dose and no E-R trends were observed at Week 52
- These results indicate that the deucravacitinib 6 mg QD dose provides an optimal benefit-risk assessment in patients with moderate to severe psoriasis

References

1. Burke JR, et al. *Sci Transl Med*. 2019;11:1-16. **2.** Lu X, et al. *Angew Chem Int Ed Engl*. 2020;59:13764-13776. **3.** Papp K, et al. *N Engl J Med*. 2018;379:1313-1321. **4.** Armstrong A, et al. Presented at the Annual Meeting of the American Academy of Dermatology; April 23-25, 2021.

Acknowledgments

- These clinical trials were sponsored by Bristol Myers Squibb
- Writing and editorial assistance were provided by Lisa Feder, PhD, of Peloton Advantage, LLC, an OPEN Health company, Parsippany, NJ, USA, funded by Bristol Myers Squibb

Disclosures

- **JS, MM, AR, EC, and AC:** Employees and shareholders: Bristol Myers Squibb
- **ML:** Nothing to disclose

Presented at the American Academy of Dermatology (AAD) Annual Meeting; March 25–29, 2022; Boston, MA

Email for Jun Shen, PhD: Jun.Shen@bms.com

This poster may not be reproduced without written permission from the authors.